#2608 Hu2G10 antibody-drug conjugates selectively target cleaved-activated Trop-2 in cancer cells and show therapeutic efficacy against multiple human cancers

Saverio Alberti, MD, PhD[1,2*]; Marco Trerotola, PhD[3]; Antonino Moschella, MD[1]; Raffaella Giavazzi[4]; Emanuela Guerra, PhD[5]

www.mediterranea-theranostic.com/en/

[1]Unit of Medical Genetics, Department of Biomedical Sciences - BIOMORF, University of Messina, Messina, Italy [2]Mediterranea Theranostic Srl, Lanciano, Italy [3]Laboratory of Cancer Pathology, CAST, and Department of Medical, Oral and Biotechnological Sciences, "G. d'Annunzio" University of Chieti-Pescara, Chieti, Italy [4]Laboratory of Cancer Metastasis Therapeutics, Istituto di Ricerche Farmacologiche Mario Negri IRCCS, Milano, Italy [5]Department of Medicine and Aging Sciences,nSection of Biomorphology, "G. d'Annunzio" University of Chieti-Pescara, Chieti, Italy *Correspondence: alberti.saverio@gmail.com

ABSTRACT

Trop-2 is a transmembrane calcium signal transducer, which activates growth-signaling networks that converge on Akt, ERK, Cyclin D1, NFkB (1). Trop-2 cleavage by ADAM10 was shown to be an activator switch for induction of cancer growth and metastatic diffusion (2). We devised a strategy to obtain monoclonal antibodies (mAbs) capable of recognizing ADAM10-exposed sites in cancer cells. We succeeded at obtaining mAbs that efficiently bound Trop-2 expressing cancer cells and were able to inhibit cell growth in vitro. We humanized the 2G10 mAb as the IgG1/k Hu2G10 antibody. Hu2G10 binds cancer-specific, cleaved/activated Trop-2 with Kd <10-12 M. On the other end, it binds uncleaved/wtTrop-2 in normal cells with Kd 3.16x10-8 M, thus promising an unprecedented therapeutic index in patients. Antibody-drug conjugates (ADC) of Hu2G10 were obtained using stable linkers, multiple conjugation chemistries and different payloads, among them topoisomerase I inhibitors, bleomycin, RNA polymerase inhibitor, MMAE and calicheamicin. Hu2G10-ADC selectively killed Trop-2-expressing cells in vitro. Dose-response curves were obtained for the BxPC-3 pancreatic cancer, DU-145 prostate cancer, HT29 colon cancer cell lines and KM12SM/Trop2 colon cancer transfectants, reaching EC50 values as low as 5.05x10-10 M. Hu2G10-ADC drove high anticancer effectiveness on cell- and patient-derived xenografts from colon, pancreas, prostate and ovarian cancer. Effectiveness followed a gradient of payload potency, with correspondingly high therapeutic indexes. Consistently, neither detectable toxic effects nor weight loss were observed in any subgroup of treated xenotransplant-bearing mice. Taken together these findings candidate anti-Trop-2 Hu2G10-ADC for best-in-class anticancer therapy.

(1) Guerra E. et al., The Trop-2 signalling network in cancer growth. Oncogene 32, 1594-1600 (2013).

(2) Trerotola M. et al., Trop-2 cleavage by ADAM10 is an activator switch for cancer growth and metastasis. Neoplasia 23, 415-428 (2021).

BACKGROUND

Trop-2 is a type-I transmembrane protein encoded by the tumor-associated calcium signal transducer 2 (TACSTD2/TROP2) gene, which is widely overexpressed in the majority of human carcinomas. Here, Trop-2 is cleaved by ADAM10 to drive tumor cell growth and metastasis, through the activation of a dormant, ubiquitously-expressed signaling super-complex and downstream effectors.

The Hu2G10 mAb targets this cancer vulnerability by binding with high efficiency and specificity the cleaved/activated Trop-2 (c-Trop-2).

Multiple Hu2G10 antibody-drug conjugates targeting c-Trop-2 were obtained and assayed against human cancers in vitro and in vivo.

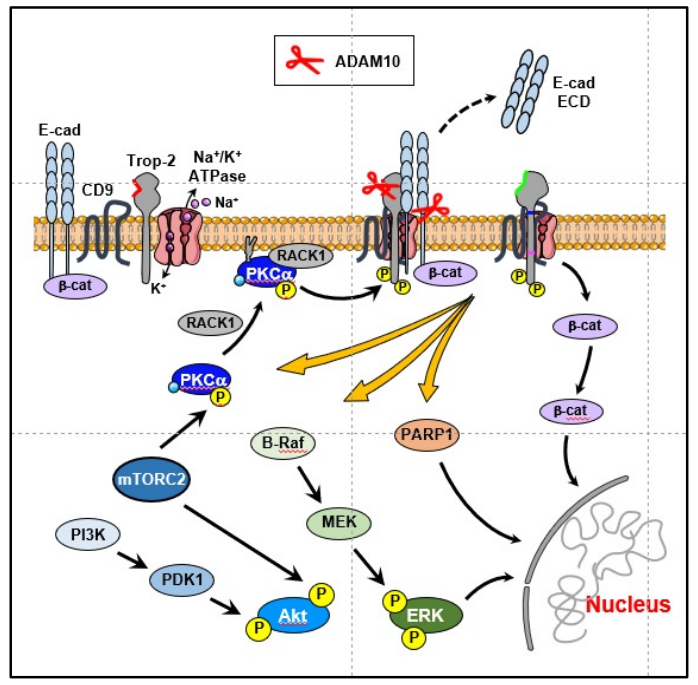
ADAM10
E-cad ECD
E-cad
CD9
Trop-2
Na+/K+ ATPase
Na+
K+
β-cat
RACK1
PKCα
P
PI3K
PDK1
mTORC2
B-Raf
MEK
PARP1
Akt
ERK
Nucleus

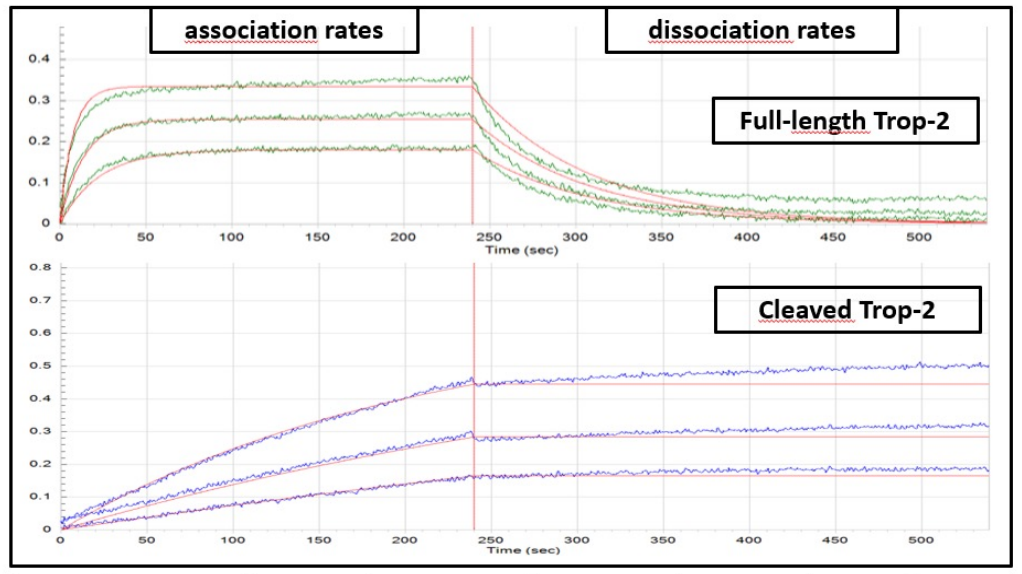
association rates
dissociation rates
Full-length Trop-2
Cleaved Trop-2
Time (sec)

Ligand	Analyte	Conc. (nM)	KD (M)	k on (1/Ms)	k dis (1/s)	χ^2	R^2
Hu 2G10	T2 –full length	250-62.5	3.16E-08	4.68E+05	1.48E-02	1.2872	0.9682
Hu 2G10	T2-Cleaved	25-6.25	<1.0E-12	1.90E+05	<1.0E-07	1.4568	0.9772

Top: cartoon depicting Trop-2 signaling cascade activation by ADAM10 cleavage between R87 and T88 in the first loop of Trop-2 extracellular domain (adapted from JCO 2023 41:4688).

Left: Hu2G10 absolute affinity to full length and cleaved Trop-2 as measured by bio-layer interferometry-based label-free binding. Association/dissociation kinetics are shown (Mol Cancer Ther 2023 22:790).

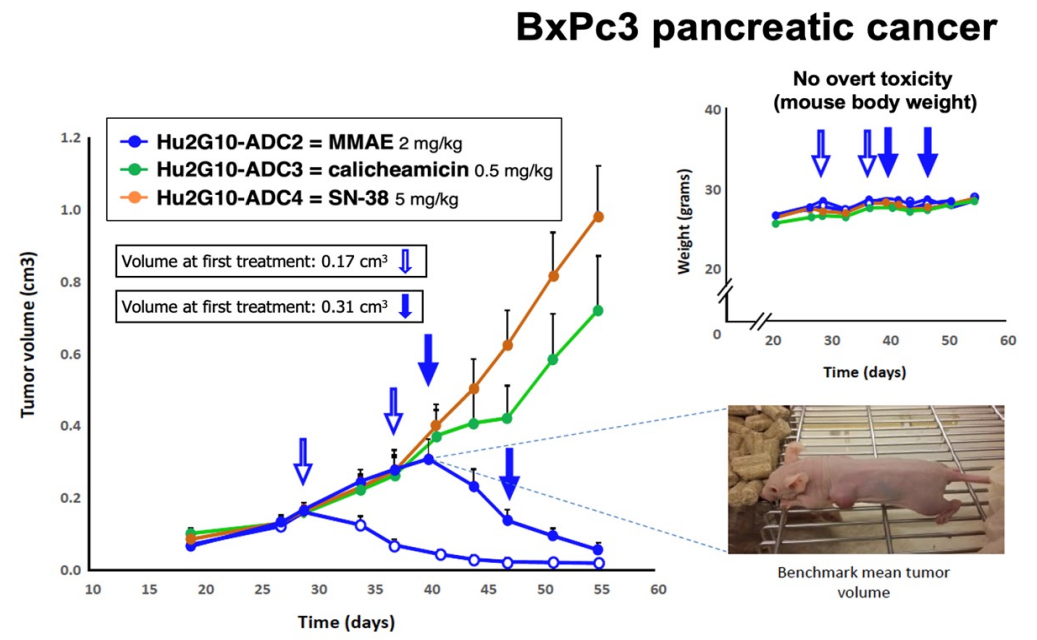
BxPc3 pancreatic cancer
Hu2G10-ADC2 = MMAE 2 mg/kg
Hu2G10-ADC3 = calicheamicin 0.5 mg/kg
Hu2G10-ADC4 = SN-38 5 mg/kg
Volume at first treatment: 0.17 cm³
Volume at first treatment: 0.31 cm³
Tumor volume (cm3)
Time (days)
No overt toxicity (mouse body weight)
Weight (grams)
Benchmark mean tumor volume

Figure 4. In vivo anticancer activity of Hu2G10 ADCs. Athymic mice subcutaneously (SC) injected with BxPC-3 pancreatic cancer cells were randomized (n=16 per group) and treated as indicated (arrows). Mice in the control groups received an irrelevant isotype-matched mAb. Tumor volumes and body weights were plotted against time. The inset shows a mouse bearing a 0.3 cm^3 tumor. Error bars: +SEM

RESULTS

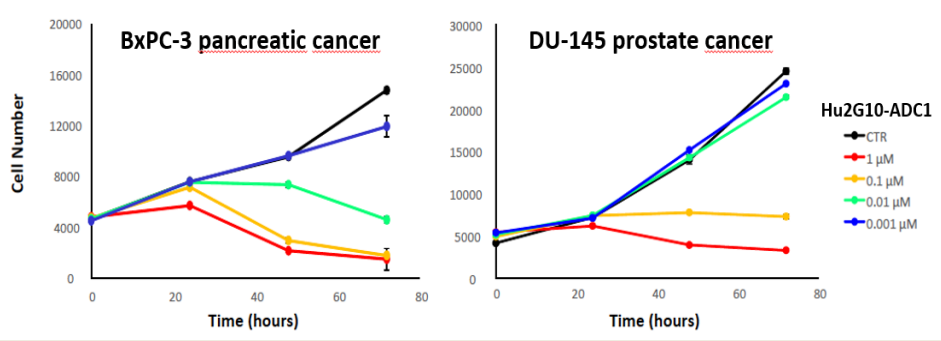
BxPC-3 pancreatic cancer
DU-145 prostate cancer
Cell Number
Time (hours)
Hu2G10-ADC1
CTR
1 μM
0.1 μM
0.01 μM
0.001 μM

Figure 1. Time course of Hu2G10-ADC1 toxicity on c-Trop-2 expressing human cancer cells (payload=α-amanitin). Bars: +SEM

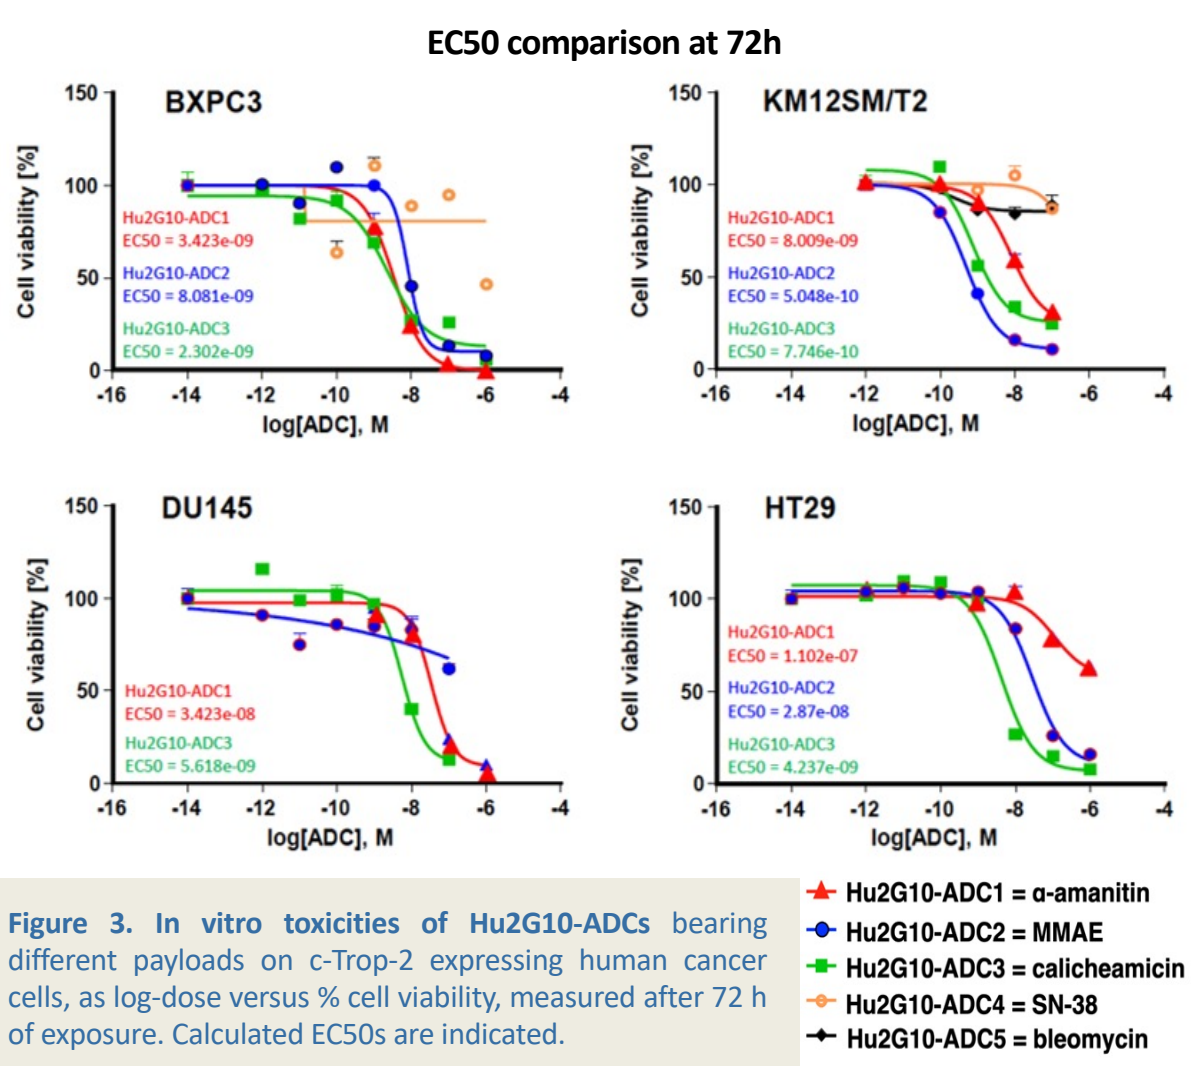
EC50 comparison at 72h
BXPC3
Hu2G10-ADC1 EC50 = 3.423e-09
Hu2G10-ADC2 EC50 = 8.081e-09
Hu2G10-ADC3 EC50 = 2.302e-09
KM12SM/T2
Hu2G10-ADC1 EC50 = 8.009e-09
Hu2G10-ADC2 EC50 = 5.048e-10
Hu2G10-ADC3 EC50 = 7.746e-10
DU145
Hu2G10-ADC1 EC50 = 3.423e-08
Hu2G10-ADC3 EC50 = 5.618e-09
HT29
Hu2G10-ADC1 EC50 = 1.102e-07
Hu2G10-ADC2 EC50 = 2.87e-08
Hu2G10-ADC3 EC50 = 4.237e-09
Cell viability [%]
log[ADC], M
Hu2G10-ADC1 = α-amanitin
Hu2G10-ADC2 = MMAE
Hu2G10-ADC3 = calicheamicin
Hu2G10-ADC4 = SN-38
Hu2G10-ADC5 = bleomycin

Figure 3. In vitro toxicities of Hu2G10-ADCs bearing different payloads on c-Trop-2 expressing human cancer cells, as log-dose versus % cell viability, measured after 72 h of exposure. Calculated EC50s are indicated.

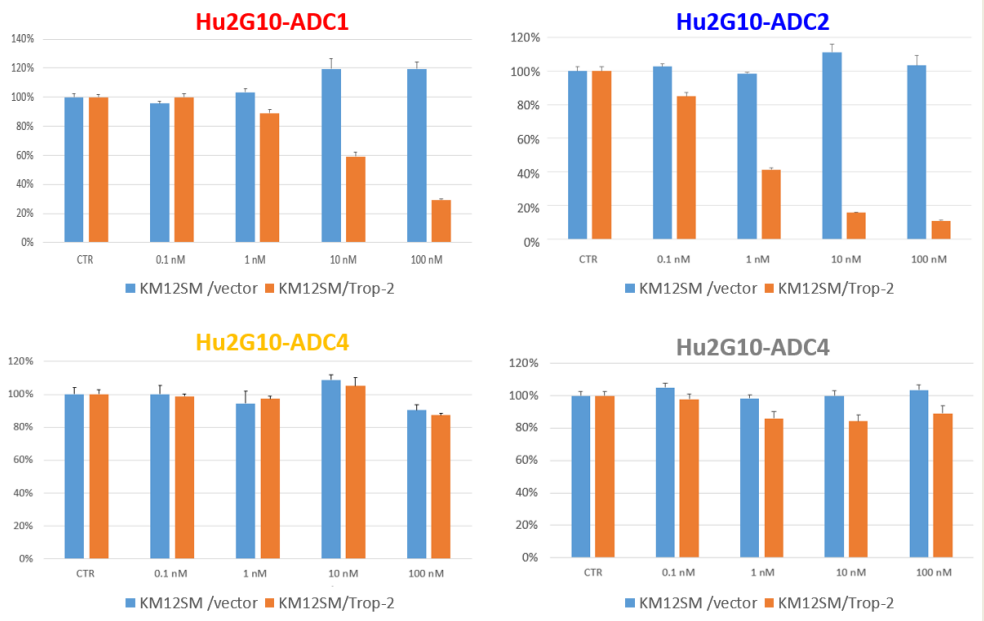
Hu2G10-ADC1
Hu2G10-ADC2
Hu2G10-ADC4
Hu2G10-ADC4
CTR
0.1 nM
1 nM
10 nM
100 nM
KM12SM /vector
KM12SM/Trop-2

Figure 2. No off-target toxicities of Hu2G10-ADCs in vitro. KM12SM/Trop-2 and /vector only mock transfectants were assayed in parallel. Normalized values are shown. Bars: +SEM

SUMMARY OF Hu2G10-ADCs IN VITRO TOXICITIES

	Hu2G10-ADC1 α-amanitin, RNA polymerase inhibitor	Hu2G10-ADC2 MMAE, microtubule disrupting agent	Hu2G10-ADC4 SN-38 topoisomerase I inhibitor	Hu2G10-ADC5 Bleomycin-DNA damaging agent
DU-145 Prostate cancer	3.42×10^{-8} M -7.47	Not Found (> 1×10^{-6} M)		
HT29 Colon cancer	1.10×10^{-7} M -6.96	2.87×10^{-8} M -7.54		
BXPC3 Pancreatic cancer	3.42×10^{-9} M -8.47	8.08×10^{-9} M -8.09	Not found	
KM12SM/Trop-2 Colon cancer transfectants	8.00×10^{-9} M -8.10	5.05×10^{-10} M -9.30	2.32×10^{-4} M -3.63	2.11×10^{-2} M -1.68
KM12SM/vector Colon cancer mock transfectants	Not found	Not found	Not found	Not found

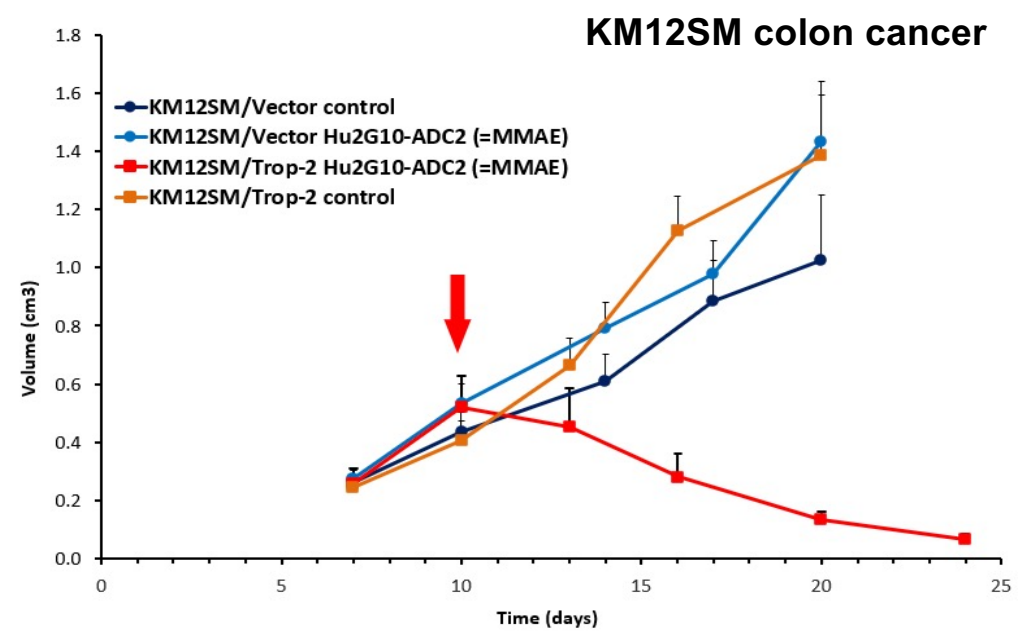
KM12SM colon cancer
KM12SM/Vector control
KM12SM/Vector Hu2G10-ADC2 (=MMAE)
KM12SM/Trop-2 Hu2G10-ADC2 (=MMAE)
KM12SM/Trop-2 control
Volume (cm3)
Time (days)

Figure 5. In vivo anticancer activity and specificity of Hu2G10-ADC. Athymic mice SC injected with cancer cell transfectants (n=16 per group) were treated with 4mg/kg of the Hu2G10-ADC2 or with an equal dose of an irrelevant isotype-matched mAb as control. KM12SM/Trop-2 and /vector only (mock) transfectants were tested in parallel. Tumor volumes were plotted against time. Arrow: time at treatment. Error bars: +SEM

CONCLUSIONS

Hu2G10-ADCs showed specific killing of c-Trop-2 expressing cancer cells in vitro. Among the payloads that were tested, monomethyl auristatin E (MMAE) showed the highest potency, with the notable exception of prostate cancer. No off-target toxicities were observed.

MMAE-based Hu2G10-ADCs showed high efficacy and specificity in vivo against cancer cells expressing c-Trop-2. No adverse effects were observed in treated mice.

Our findings candidate the anti-c-Trop-2 Hu2G10 for best-in-class ADC anticancer therapy.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com